Novel Conditionally Active Tetravalent B7-H3 x CD3 T-cell Engagers Targeting Solid Tumors

Ana Paula Cugnetti, Haizhen Liu, Patricia McNeeley, Kathryn Woodard, Cathy Chang, Gerhard Frey, William J. Boyle, and Jay M. Short

BioAtla, 11085 Torreyana Road, San Diego, CA 92121

Abstract number 6340

INTRODUCTION

B7-H3, also known as CD276, is an immune-checkpoint molecule and a type 1 transmembrane protein that exerts a variety of biological functions. In cancer biology, the overexpression of B7-H3 is correlated with tumor cell invasion and metastasis, decreased T-cell infiltration and function, poor prognosis, and resistance to therapy. As B7-H3 is expressed in multiple solid tumors and plays an important role in modulating anti-tumor immune response as well as promoting cancer cell formation, it became an important target for the development of antibody-based immunotherapy. Many forms of bispecific antibodies targeting B7-H3 have shown antitumor response in preclinical studies. However, bispecific T-cell engagers (TCE) targeting B7H3 and CD3 have been correlated with cytokine release syndrome and hepatic adverse events, a common toxicity observed using TCEs which has limited the development of this immunotherapeutic in clinic.

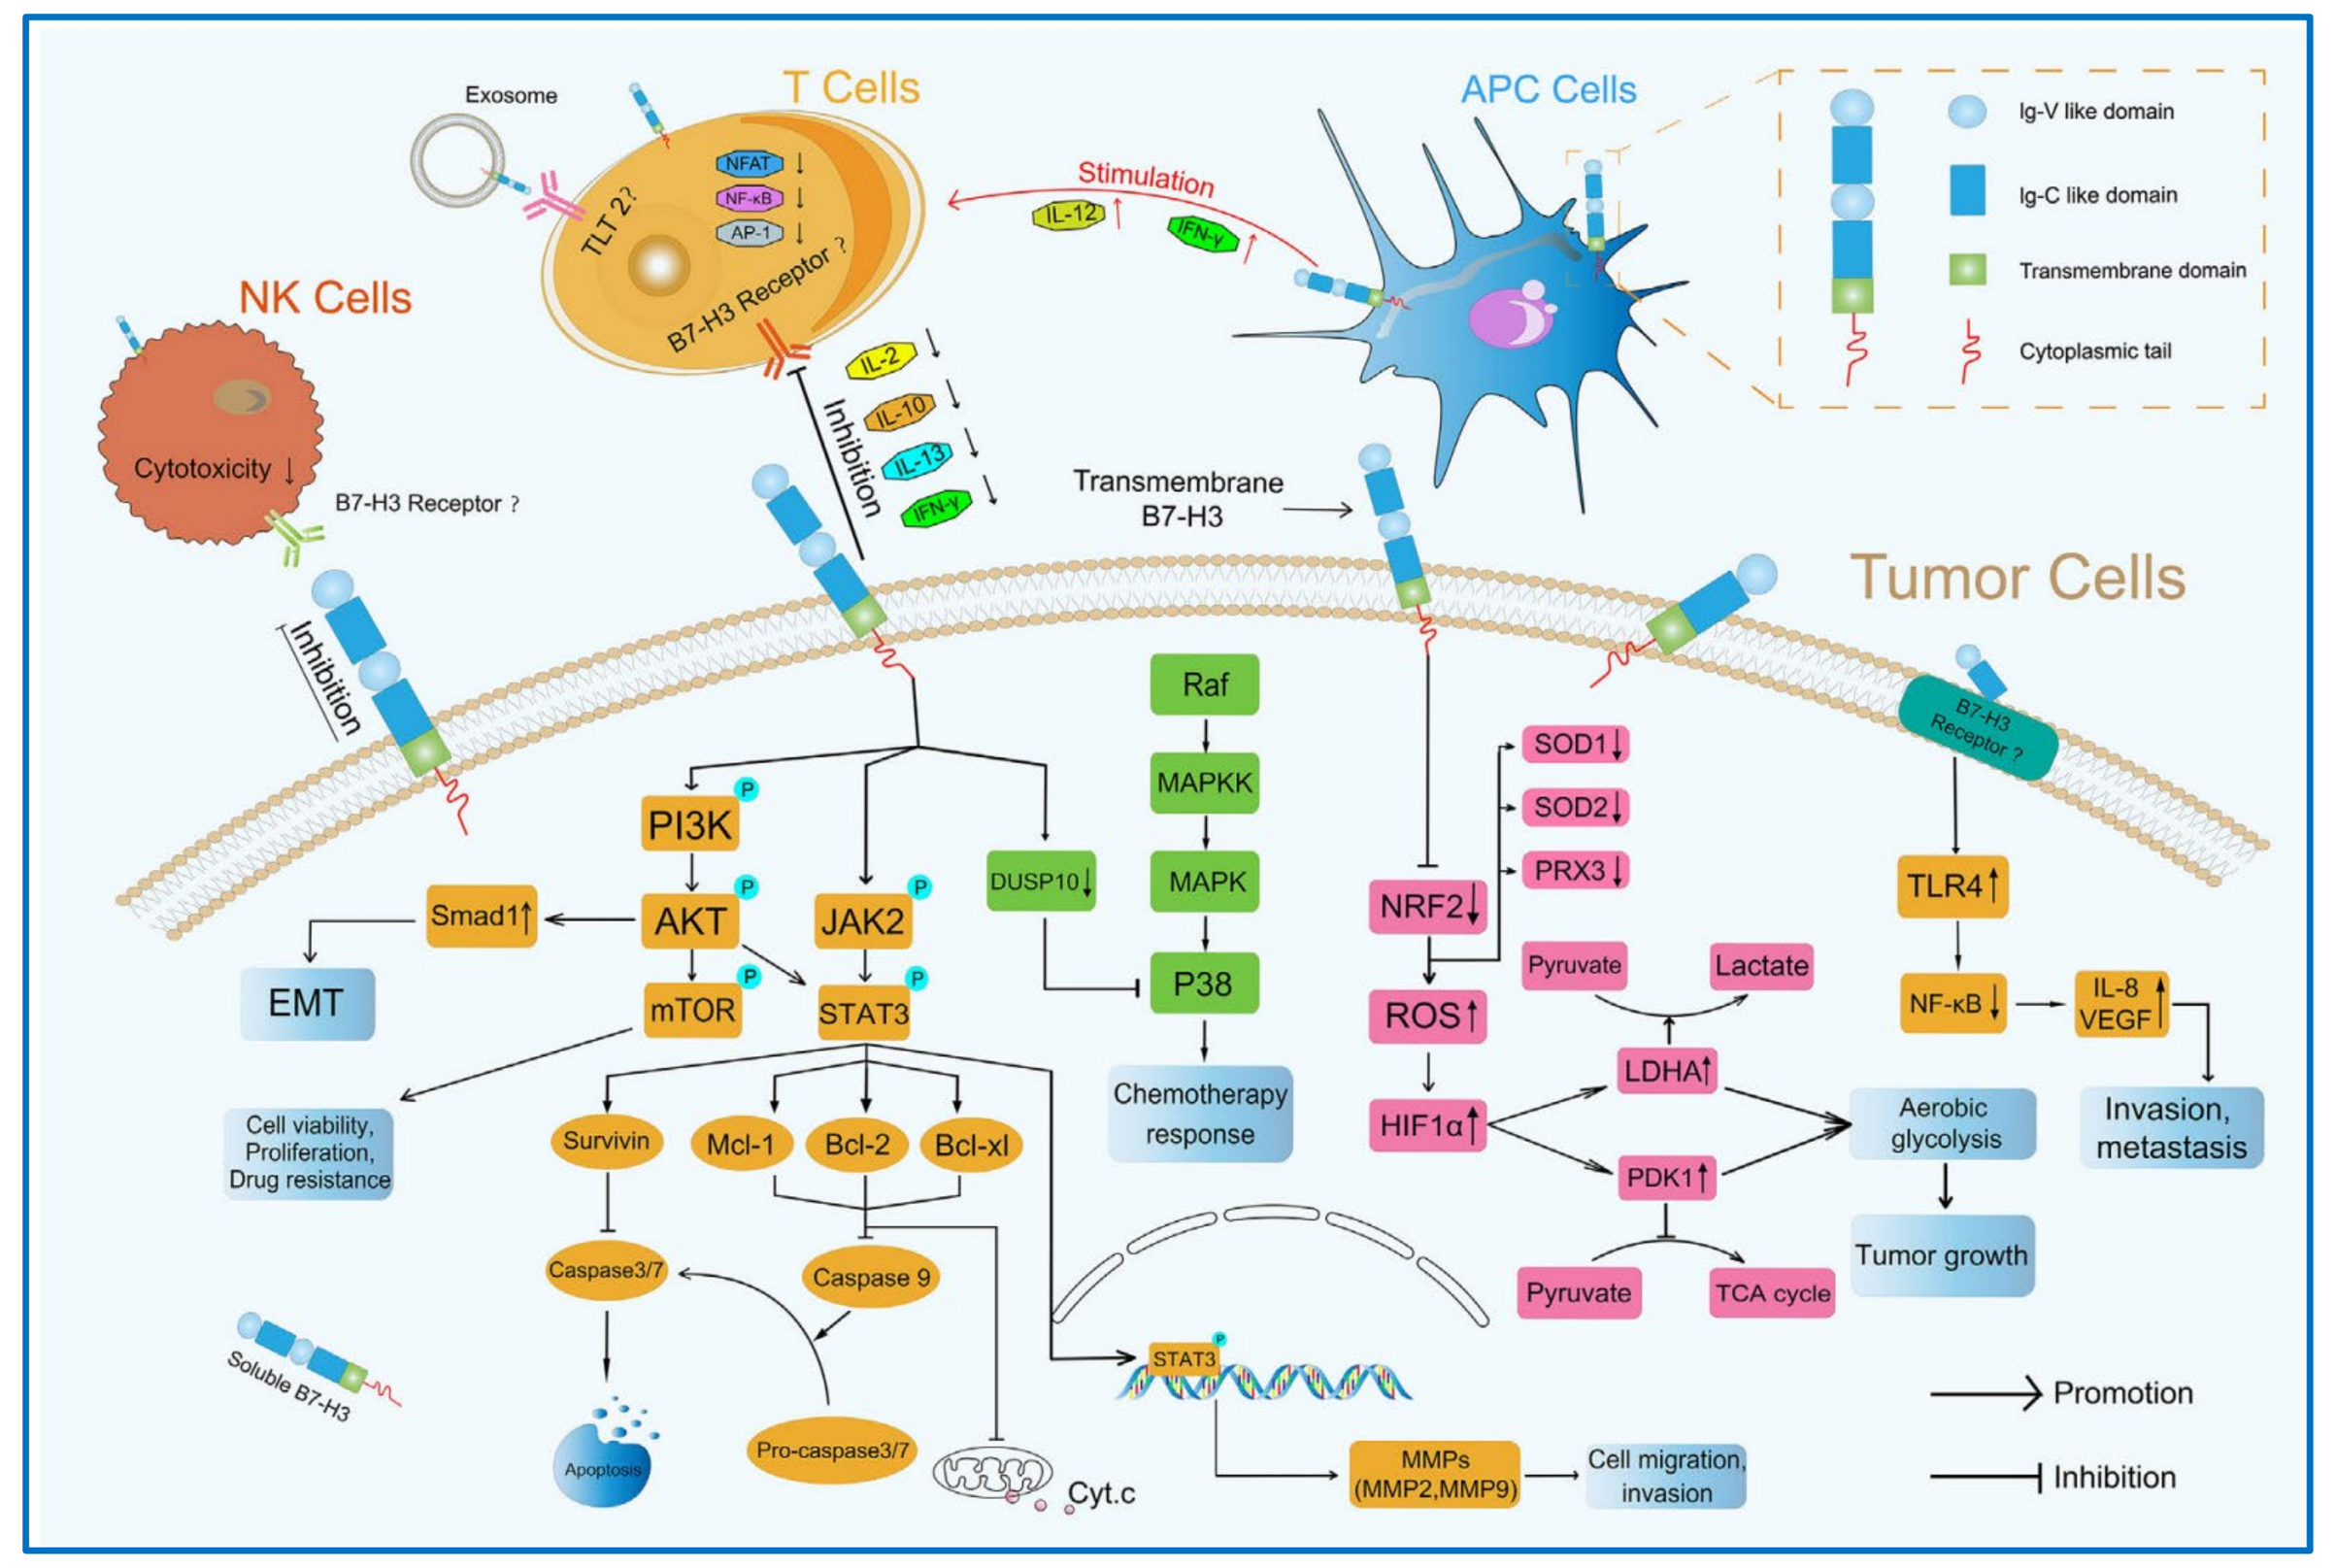
T Cells
APC Cells
NK Cells
Tumor Cells
Exosome
Stimulation
Inhibition
Transmembrane B7-H3
Cytotoxicity
B7-H3 Receptor ?
Ig-V like domain
Ig-C like domain
Transmembrane domain
Cytoplasmic tail
PI3K
AKT
mTOR
JAK2
STAT3
Smad1
EMT
Raf
MAPKK
MAPK
P38
DUSP10
Chemotherapy response
NRF2
ROS
HIF1α
SOD1
SOD2
PRX3
Pyruvate
Lactate
LDHA
PDK1
TCA cycle
TLR4
NF-κB
IL-8 VEGF
Aerobic glycolysis
Invasion, metastasis
Tumor growth
Survivin
Mcl-1
Bcl-2
Bcl-xl
Caspase3/7
Caspase 9
Pro-caspase3/7
Cyt.c
MMPs (MMP2,MMP9)
Cell migration, invasion
Cell viability, Proliferation, Drug resistance
Soluble B7-H3
Promotion
Inhibition

Figure 1[a]: Roles of B7-H3 in tumor microenvironment (TME). B7-H3 can affect the progression of tumors through immune-dependent and nonimmune pathways.

[a] Zhou W-T and Jin W-L (2021) B7-H3/CD276: An Emerging Cancer Immunotherapy. Front. Immunol. 12:701006.

RATIONALE

Conditionally Active Biologic (CAB) technology[1] is a proprietary platform that generates antibodies that have little to no binding to their target antigen in healthy tissue (normal alkaline microenvironment). However, in acid conditions that mirror the tumor microenvironment (high glycolysis) the binding of these antibodies to their target molecule is strong. Using the CAB technology, we developed BA3142, a DualCAB T-cell engager targeting B7H3 and CD3. BA3142 binds with high affinity to both B7-H3 and CD3 molecules under conditions that mimic the acidic tumor microenvironment, but with lower affinity in alkaline physiological conditions. BA3142 inhibited tumor growth of B7H3 positive human tumor xenografts *in vivo*. Toxicity of BA3142 was evaluated in a single and repeat dose finding study in non-human primate (NHP). BA3142 was well tolerated and overall safe in NHP. No BA3142-related mortality occurred, and no clinical signs of toxicity were observed. The maximum tolerated dose (MTD) of BA3142 was 25 mg/kg/dose after a single or repeat-dosing. Given the efficacious and safety profile of BA3142, we expect to yield a superior therapeutic index relative to other T-cell engagers targeting B7H3.

1. Chang H.W., Frey G., Liu H., Xing C., Steinman L., Boyle W.J., Short J.M. Proc. Natl. Acad. Sci. U.S.A. 2021 Mar. 2;118(9).

RESULTS

- Differential binding of BA3142 and Non-CAB B7H3/CD3 TCE to recombinant human B7H3 extra cellular domain and CD3 epsilon/delta heterodimer in conditions mimicking the tumor microenvironment pH, compared to conditions mimicking the normal tissue pH.

(A) Tumor Microenvironment pH **(B) Normal Physiological pH**

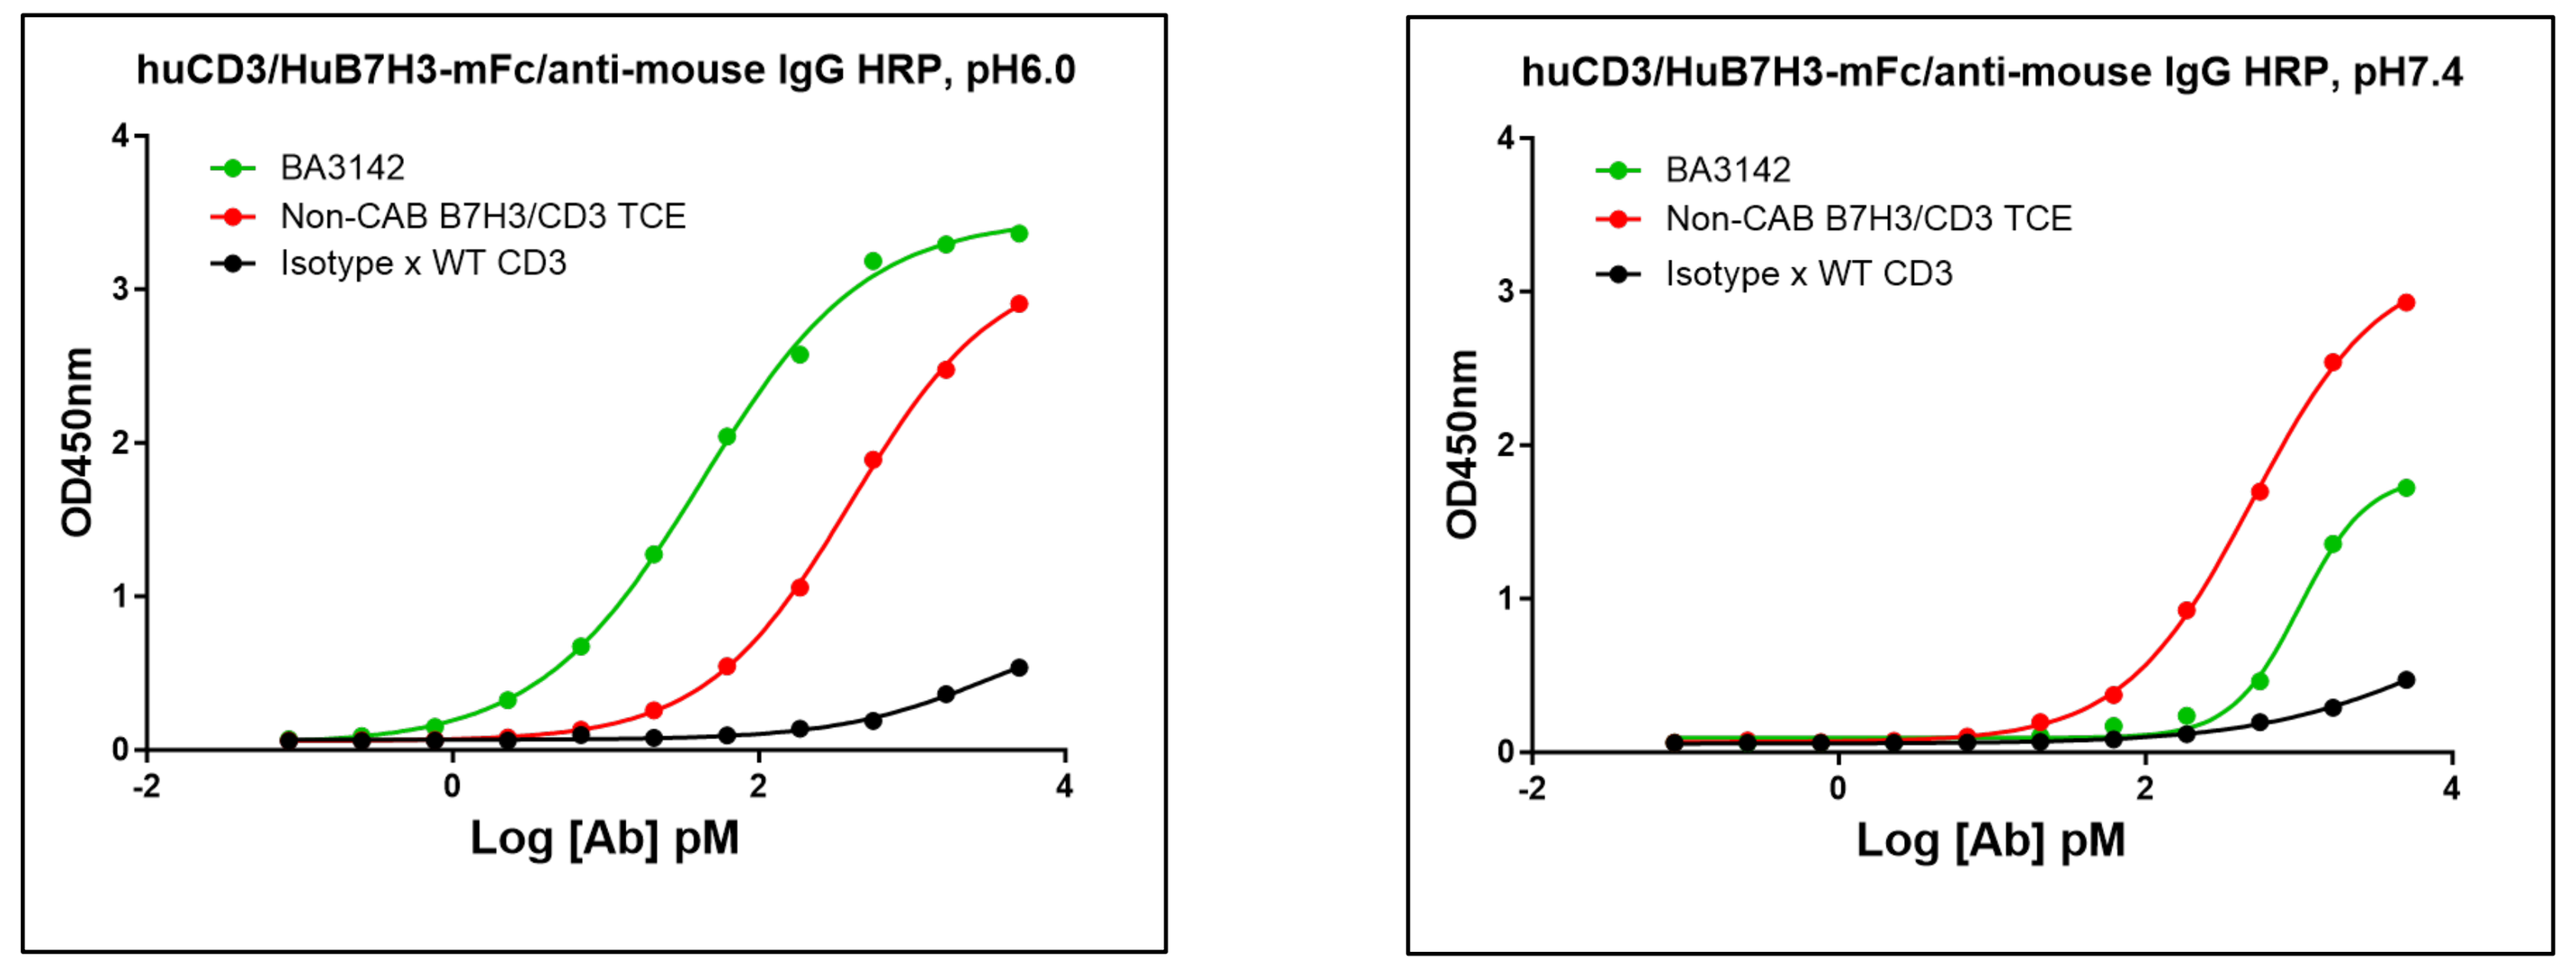
huCD3/HuB7H3-mFc/anti-mouse IgG HRP, pH6.0
huCD3/HuB7H3-mFc/anti-mouse IgG HRP, pH7.4
BA3142
Non-CAB B7H3/CD3 TCE
Isotype x WT CD3
OD450nm
Log [Ab] pM

Figure 2. Differential affinity binding pH affinity ELISA using human CD3 as capture antigen, human B7H3-mFc as detection followed by anti-mouse IgG HRP conjugated antibody.

- BA3142 is more potent in inducing T cell activation and cytotoxicity of cancer cells in tumor microenvironment pH 6.0 **(A)** and less potent in physiological pH 7.4 **(B)**.

(A) Tumor Microenvironment pH **(B) Normal Physiological pH**

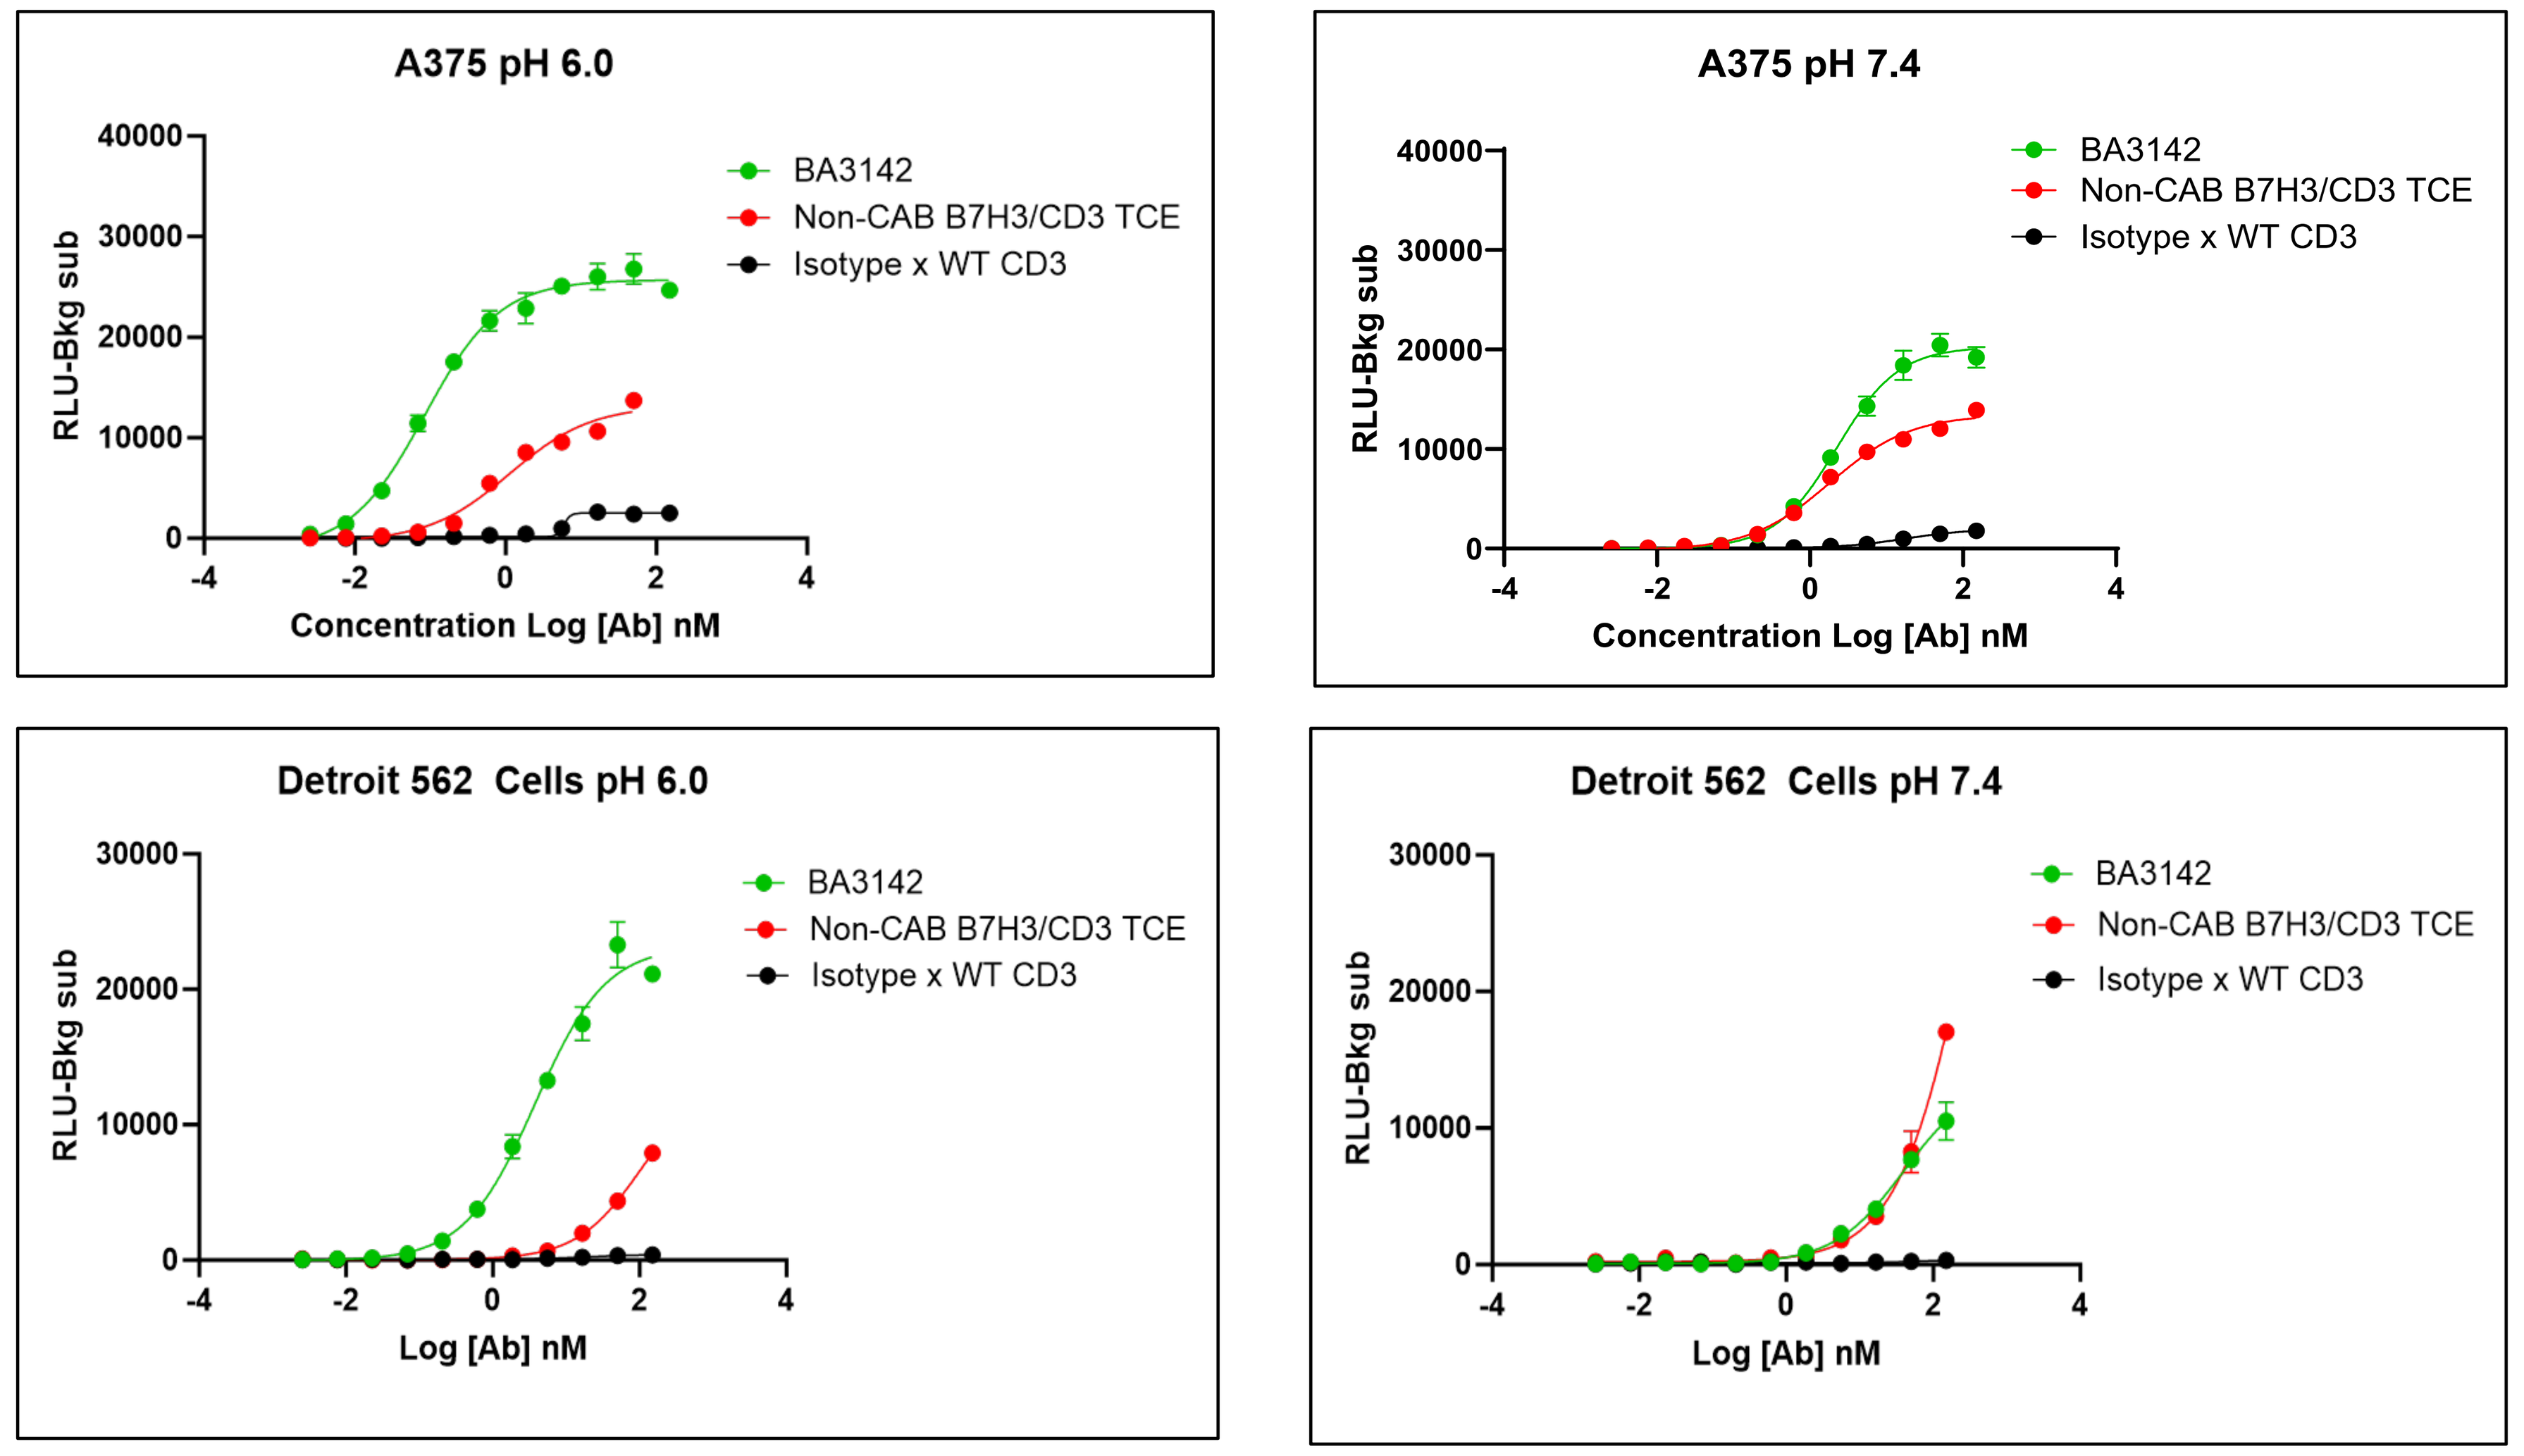
A375 pH 6.0
A375 pH 7.4
Detroit 562 Cells pH 6.0
Detroit 562 Cells pH 7.4
BA3142
Non-CAB B7H3/CD3 TCE
Isotype x WT CD3
RLU-Bkg sub
Concentration Log [Ab] nM
Log [Ab] nM

(C) B7H3/CD3 TCEs EC50 values

Bispecific Antibodies	A375 Cells EC50 (nM)		Detroit 562 Cells EC50 (nM)	
	pH6.0	pH7.4	pH6.0	pH7.4
Non-CAB B7H3/CD3 TCE	1.13	1.83	N/A	N/A
BA3142	0.09	2.25	3.8	39.7
Isotype x WT CD3	N/A	N/A	N/A	N/A

Figure 3: **B7H3 TCEs induce T-cell activation *in vitro*.** A375 and Detroit 562 cells were cocultured with TCR/CD3 Jurkat effector cells that express a luciferase reporter driven by NFAT-response element (Promega T-cell Activation Assay). Co-cultures were incubated in the presence of B7H3/CD3 TCEs at pH 6.0 and pH 7.4. **(A)** Tumor microenvironment pH, **(B)** Normal Physiological pH and **(C)** EC50 values.

RESULTS

- BA3142 dosed at 2mg/kg led to significant tumor regression in Detroit 562 and A375 humanized mouse model of human pharyngeal and melanoma cancers, respectively. The *in vivo* anti-tumor activity of BA3142 was comparable to the Non-CAB B7H3/CD3 TCE.

(A) Detroit 562 Humanized Mouse Model **(B) A375 Humanized Mouse Model**

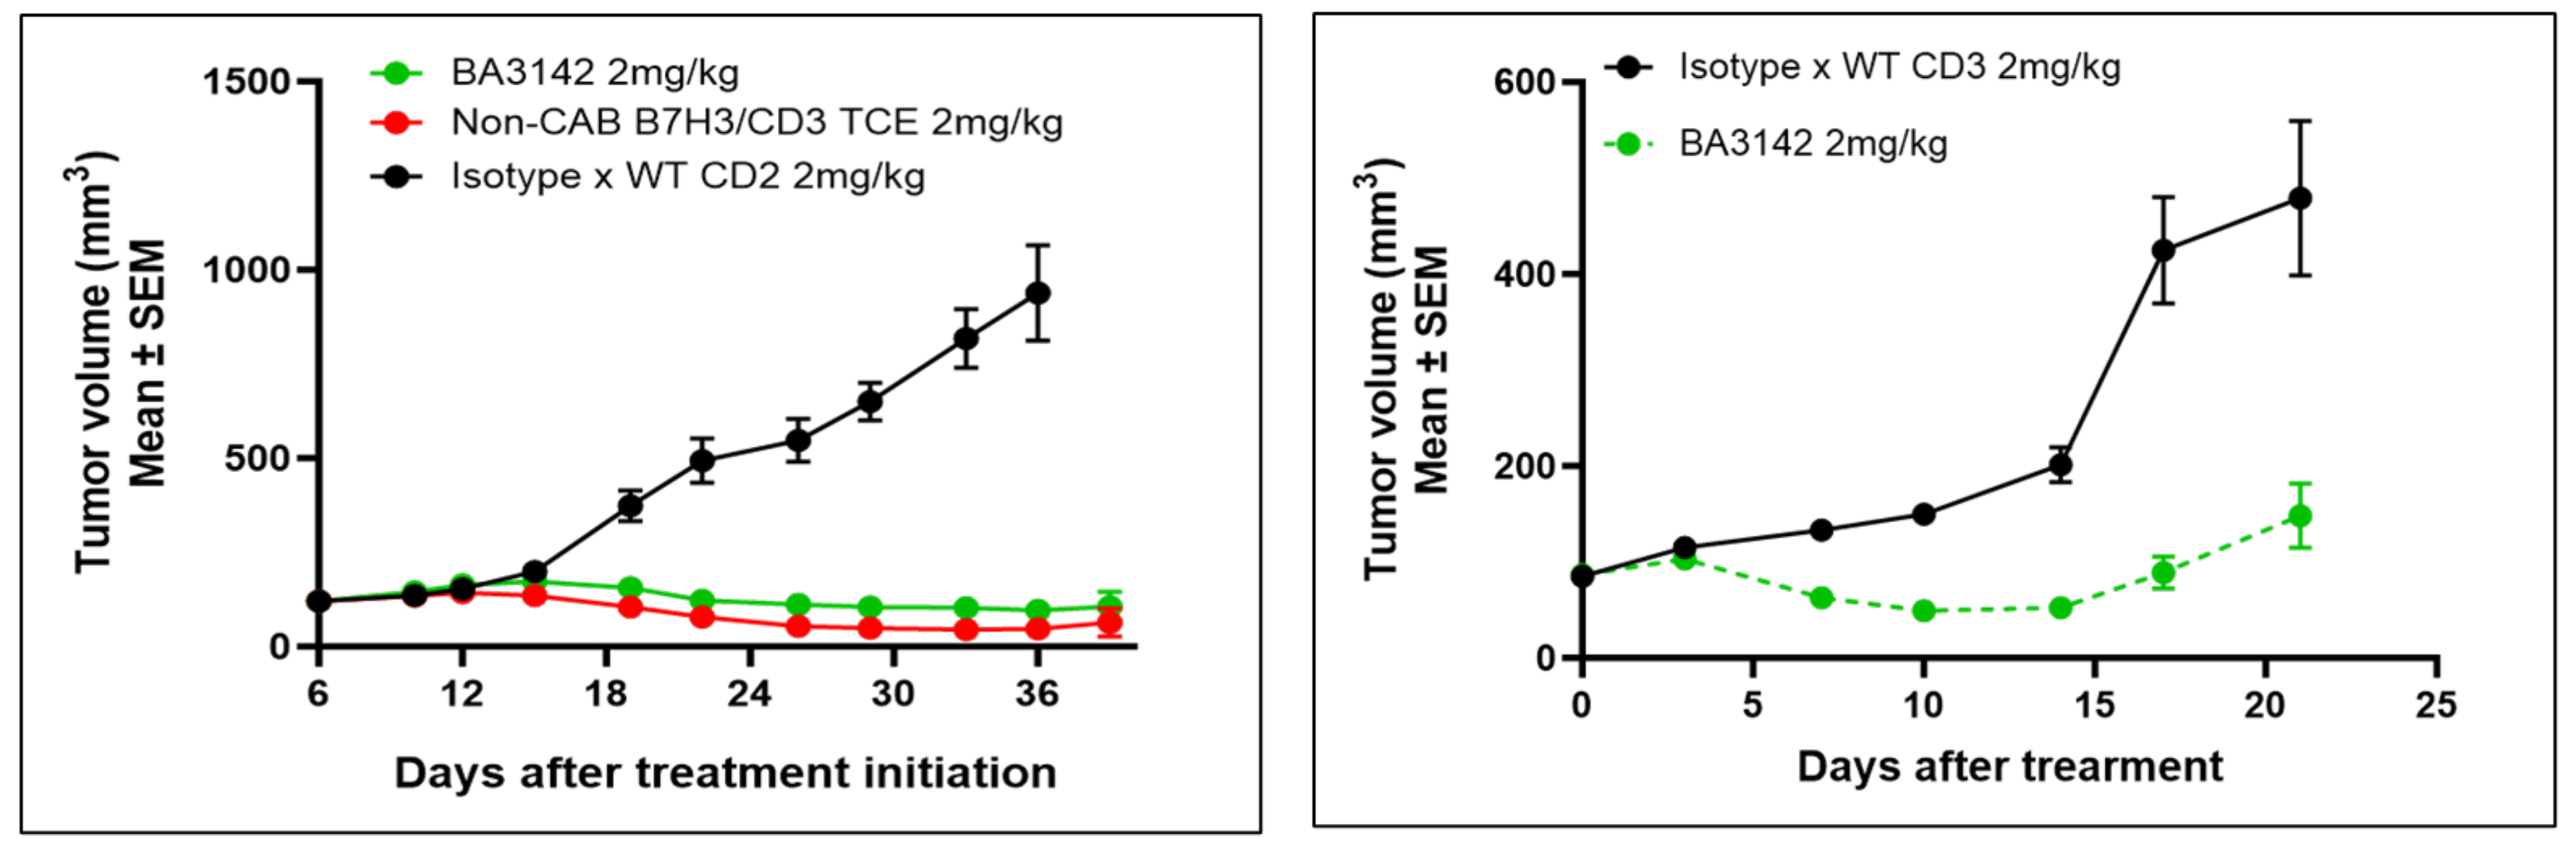
BA3142 2mg/kg
Non-CAB B7H3/CD3 TCE 2mg/kg
Isotype x WT CD2 2mg/kg
Isotype x WT CD3 2mg/kg
Tumor volume (mm³) Mean ± SEM
Days after treatment initiation
Days after trearment

Figure 4. *In vivo* efficacy studies. Triple immunodeficient mice were engrafted with human PBMCs and inoculated with Detroit 562 **(A)** or A375 **(B)** cells. Treatment was initiated when the tumor volume reached approximately 80-120 mm^3, and mice were dosed with TCEs at 2mg/kg BIW x 4 weeks.

- In NHP BA3142 was well tolerated with a MTD of 25mg/kg. Sporadic elevations of IL-6 cytokine was observed without a clear relationship with BA3142 dosing. Systemic exposure was dose-proportional with no significant gender differences in TK parameters.

(A) IL-6 levels in NHP serum **(B) BA3142 concentration time profile**

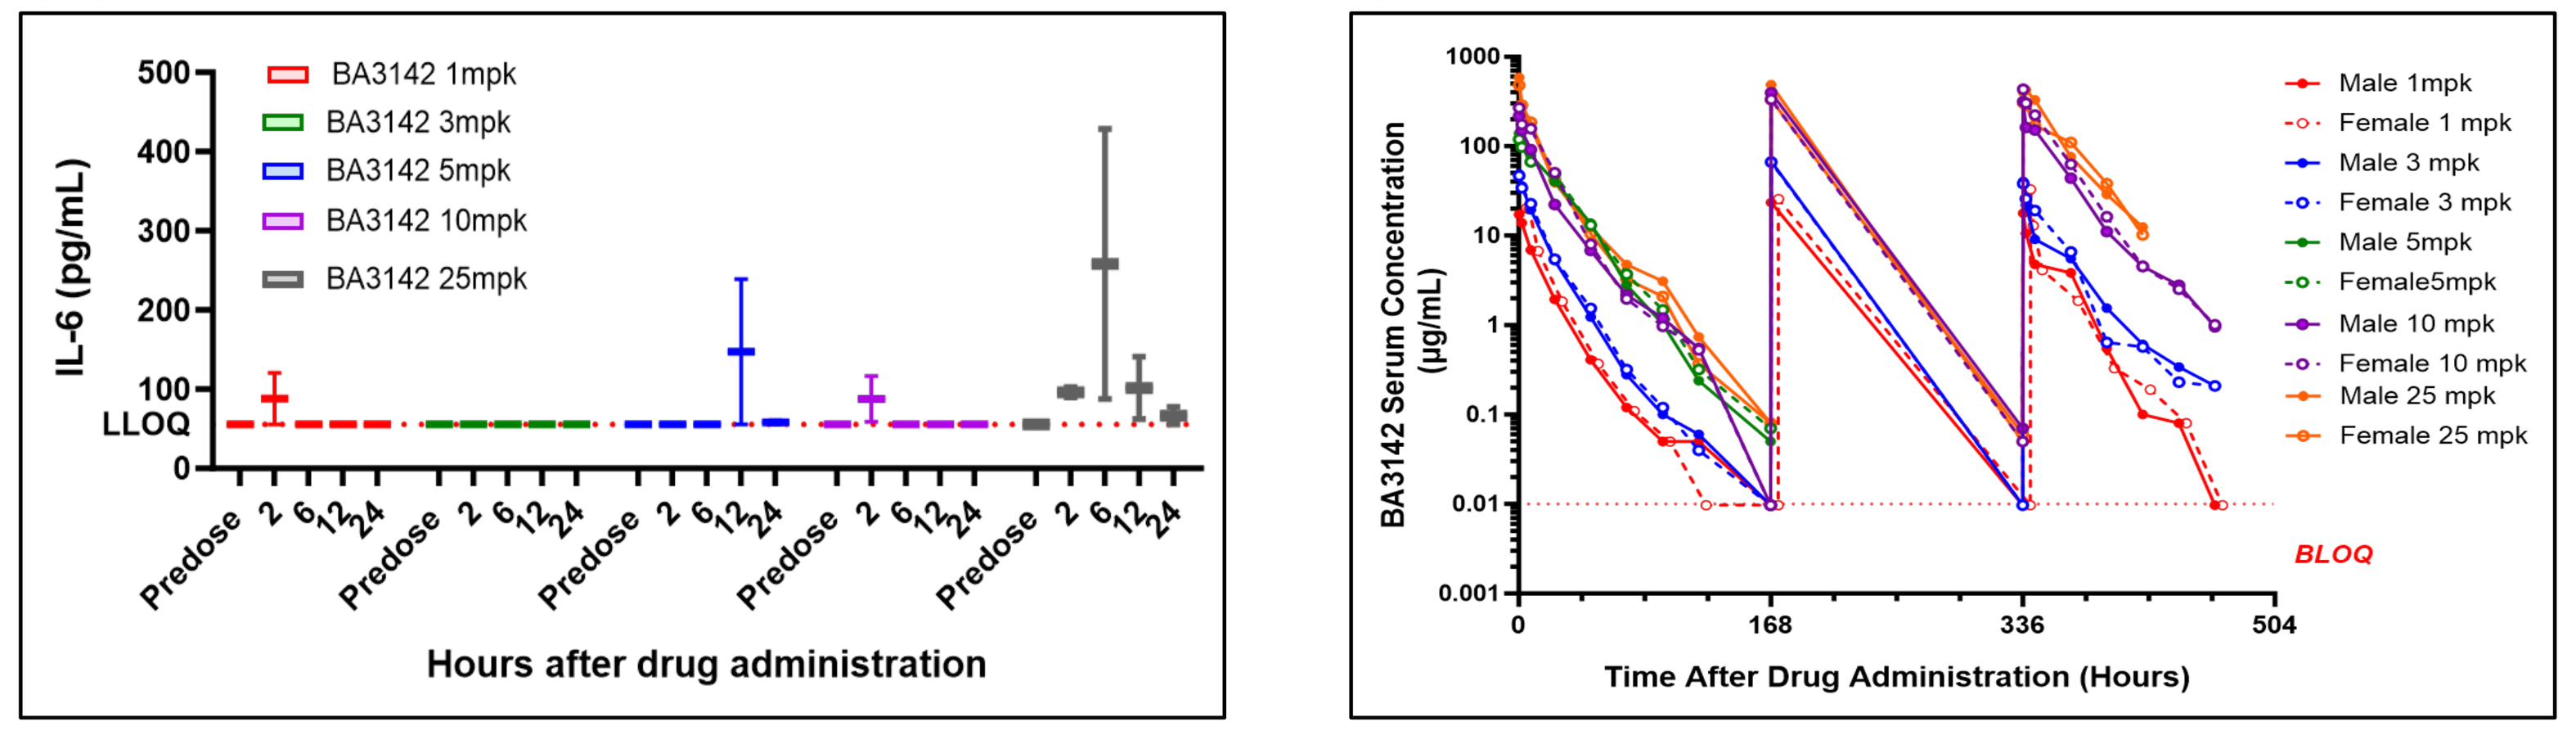
BA3142 1mpk
BA3142 3mpk
BA3142 5mpk
BA3142 10mpk
BA3142 25mpk
IL-6 (pg/mL)
LLOQ
Predose
Hours after drug administration
BA3142 Serum Concentration (µg/mL)
Time After Drug Administration (Hours)
BLOQ
Male 1mpk
Female 1 mpk
Male 3 mpk
Female 3 mpk
Male 5mpk
Female5mpk
Male 10 mpk
Female 10 mpk
Male 25 mpk
Female 25 mpk

(C) BA3142 Systemic Exposure

BA3142 Exposure in NHP – TK Parameters are shown as Mean ± SD

BA3142 Dose	C_{max} (µg/mL)	AUC_{0-168} (h*µg/mL)	Cl (mL/h/kg)	$T_{1/2}$ (h)
1mg/kg	19 ± 2	192 ± 11	5.2 ± 0.3	15 ± 2
3mg/kg	48 ± 2	520 ± 24	5.8 ± 0.3	14 ± 3
5mg/kg	129 ± 14	1280 ± 21	3.9 ± 0.1	16 ± 1
10mg/kg	243 ± 36	2970 ± 926	3.5 ± 1.1	13 ± 1
25mg/kg	795 ± 115	6780 ± 891	3.7 ± 0.5	25 ± 2

Figure 5. Toxicity study in NHP. Cynomolgus monkeys received a single or three intravenous administration of BA3142 at different doses. Serum was collected at different time points for cytokine and toxicokinetic analysis. **(A)** IL-6 cytokine levels in NHP serum, **(B)** BA3142 concentration time profile in NHP serum and **(C)** TK parameters after a single dose of BA3142.

CONCLUSIONS

- BA3142 has high affinity to human B7H3/CD3 in conditions that mimic the tumor microenvironment pH, and lower affinity and reduced binding due to PACS (1) in physiological pH.
- BA3142 TCE has comparable anti-tumor activity ***in vivo*** to the Non-CAB B7H3/CD3 TCE.
- BA3142 has safe profile in non-human primates with a MTD of 25 mg/kg without mortality or clinical signs of toxicity.
- **BioAtla has already a CAB-TCE targeting EpCAM/CD3 (BA3182) in Phase 1 clinical trial. BA3142 is the second CAB TCE targeting solid tumors, developed with the potential of widening the therapeutic index.**

AACR Annual Meeting 2024, April 5-10

www.bioatla.com
©2024 BioAtla Incorporation